#767

Safety and Preliminary Efficacy of mRNA-2752, a Lipid Nanoparticle Encapsulating mRNAs Encoding huOX40L/IL-23/IL-36γ for Intratumoral (iTu) Injection and Durvalumab (IV) in TNBC and Melanoma

Randy F. Sweis[1], Daniel Olson[1], Adil Daud[2], Ronnie Shapira-Frommer[3], Antonio Jimeno[4], Patrick Reagan[5], Shivaani Kummar[6], Raghad Abdul-Karim[7], Salomon M Stemmer[8], Meredith McKean[9], Ravit Geva[10], Ruth Perets[11], Manish R. Patel[12], Thomas Marron[13], Shilpa Gupta[14], Anupam Desai[15], Jeffrey S. Weber[16], Kim Margolin[17], Jong Chul Park[18], Sima J. Zacharek[19], Andressa S. Laino[19], Jack Pollard[19], Jing Sun[19], Josh Frederick[19], Lili Zhu[19], Xing Chen[19], Kinjal Mody[19], Natalia Lopez[19], Praveen Aanur[19], Robert S. Meehan[19], Lisa Johansen[19], Sara Lavoie[19], Khanh T. Do[19], Ryan Sullivan[18]

[1]The University of Chicago, Chicago, IL; [2]UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, CA; [3]Sheba Medical Center; Ramat Gan, Israel; [4]University of Colorado Cancer Center, Aurora, CO; [5]University of Rochester, Rochester, NY; [6]Oregon Health and Science University, Portland, OR; [7]Henry Ford Hospital, Detroit, MI; [8]Rabin Medical Center, Tel Aviv, Israel; [9]Sarah Cannon Research Institute / Tennessee Oncology, PLLC, Nashville, TN; [10]Sourasky Medical Center, Tel Aviv, Israel; [11]Rambam Medical Center and Technion – Israel Institute of Technology, Haifa, Israel; [12]Sarah Cannon Research Institute / Florida Cancer Specialists, Sarasota, FL; [13]Icahn School of Medicine at Mount Sinai, New York City, NY; [14]The Cleveland Clinic Foundation, Cleveland, OH; [15]Beth Israel Deaconess Medical Center, Boston, MA; [16]NYU Langone Health, New York, NY; [17]Saint John's Cancer Institute, Santa Monica, CA; [18]Massachusetts General Hospital, Boston, MA; [19]Moderna Inc., Cambridge, MA

SITC 37th Annual Meeting Nov. 8-12th Boston, MA

BACKGROUND

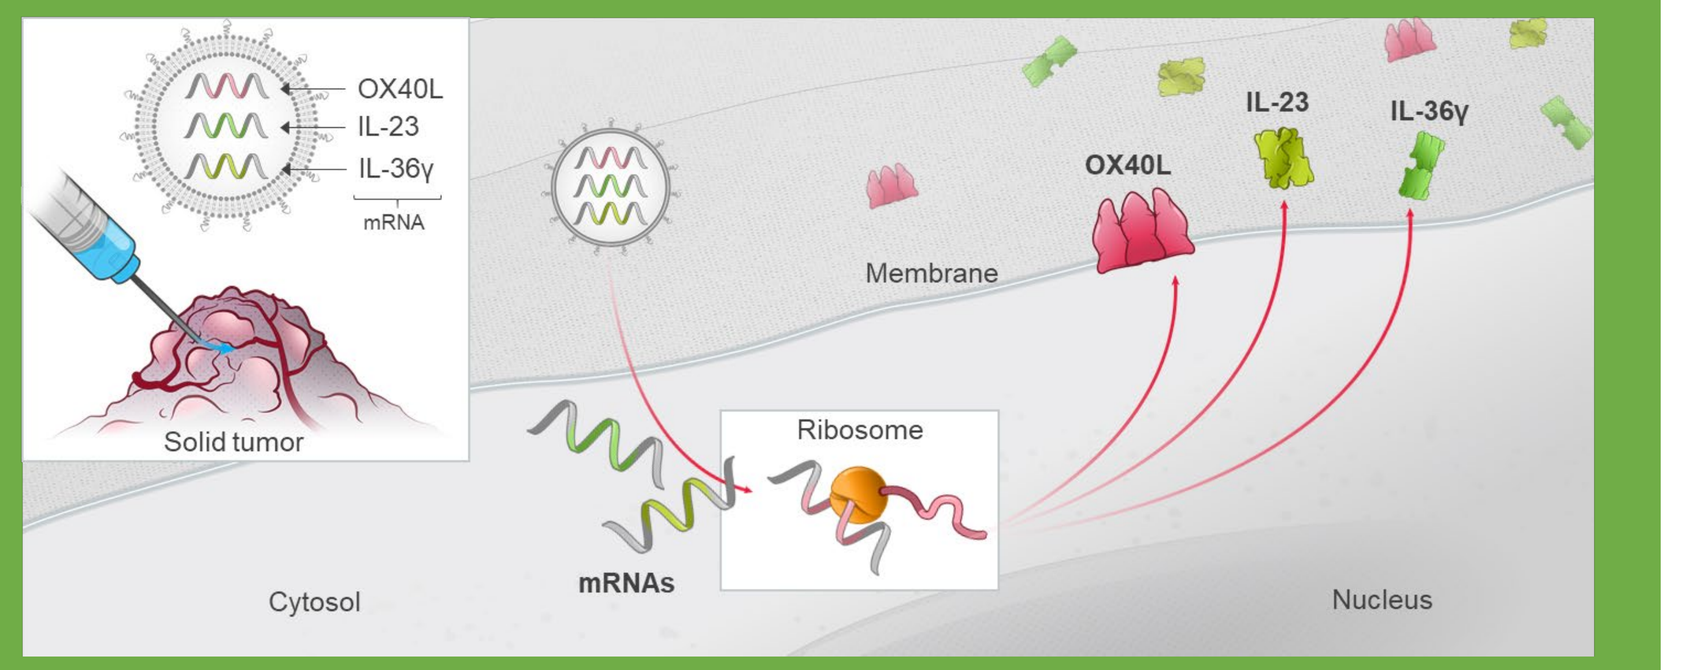

OX40L
IL-23
IL-36γ
mRNA
Solid tumor
Membrane
Ribosome
mRNAs
Cytosol
Nucleus

- mRNA-2752 is a novel mRNA-based therapeutic encoding OX40L T cell co-stimulator, IL-23 and IL-36γ pro-inflammatory cytokines
- Expression of pro-inflammatory cytokines within a treated tumor along with T-cell co-stimulation modulates the tumor microenvironment (TME) to promote a productive immune response
- Preclinical data demonstrate robust activity in syngeneic murine tumor models as a single agent and in combination with PD-L1 blockade (Hewitt et al., 2019)

OBJECTIVES

- Assess the safety and tolerability of mRNA-2752 +/- durvalumab
- Characterize the pharmacokinetics of mRNA-2752 +/- durvalumab
- Characterize protein expression from the introduced mRNAs and measure biomarkers of immune response
- Assess preliminary anti-tumor activity as measured by objective response rate (ORR), progression-free survival (PFS), and duration of response (DOR) per RECIST v1.1 for solid tumors and per Cheson for lymphomas

STUDY DESIGN

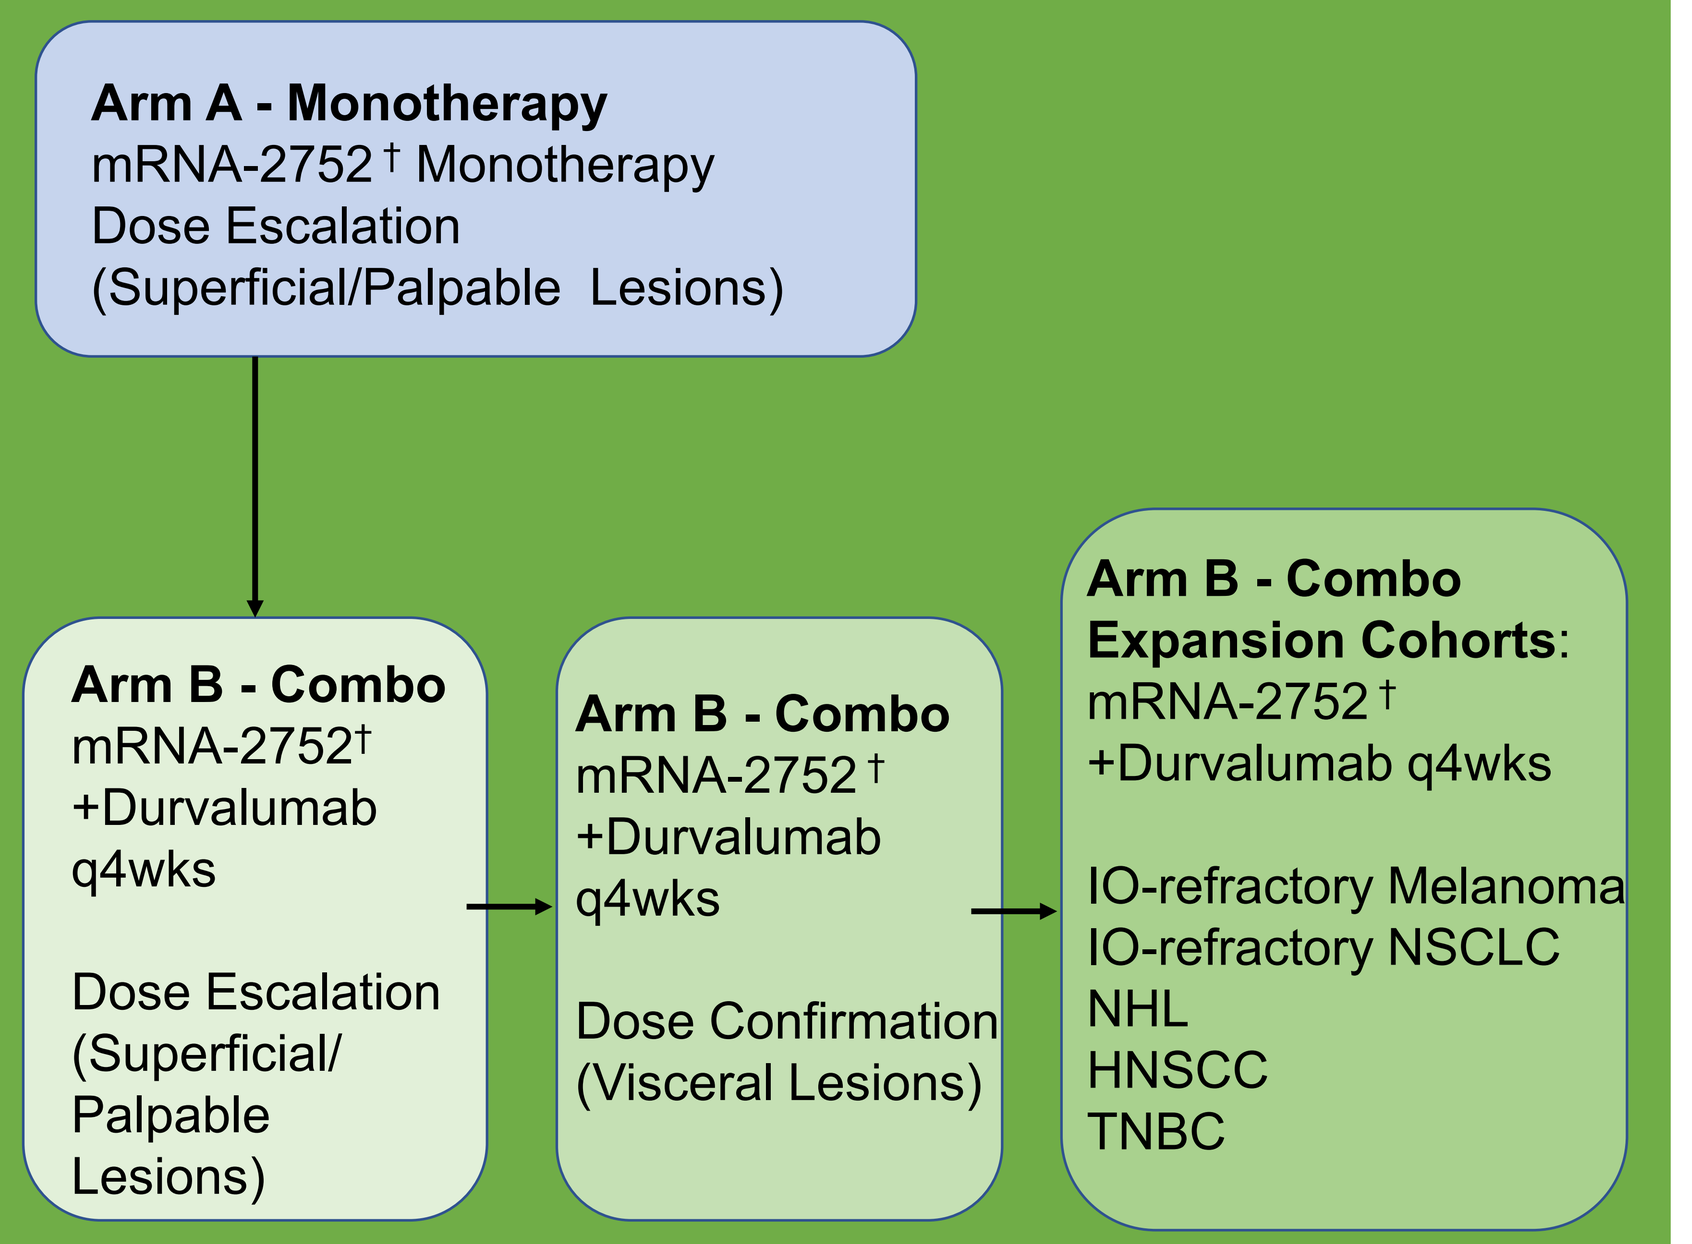

Arm A - Monotherapy
mRNA-2752† Monotherapy
Dose Escalation
(Superficial/Palpable Lesions)
Arm B - Combo
mRNA-2752†
+Durvalumab q4wks
Dose Escalation
(Superficial/ Palpable Lesions)
Arm B - Combo
mRNA-2752†
+Durvalumab q4wks
Dose Confirmation
(Visceral Lesions)
Arm B - Combo Expansion Cohorts:
mRNA-2752†
+Durvalumab q4wks
IO-refractory Melanoma
IO-refractory NSCLC
NHL
HNSCC
TNBC

† mRNA-2752 administration schedule: cycle 1(Day 1 and 15); Cycle 2-6 (Day1)

Arm C Schedule Exploration
mRNA-2752 Weekly administration
+/- Durvalumab in Neoadjuvant Melanoma

RESULTS

Demographics

Arm B (n=80 enrolled; 77 treated)	
Age(y)	
Range:	24-91
Median:	60
Sex	
Male:	27 (33.8%)
Female:	53 (66.2%)
Race:	
Caucasian	63 (78.8%)
Black/African American	7 (8.8%)
Asian	2 (2.5%)
Hispanic/Latino	3 (3.8%)
Other/Not reported	5 (6.1%)
Prior Systemic Therapies	
0-1	14 (17.5%)
2	10 (12.5%)
3	15 (18.8%)
3+	41 (51.2%)
Prior Checkpoint Inhibitor	51 (66.2%)
Atezolizumab	8 (10.4%)
Durvalumab	2 (2.6%)
Ipilimumab	12 (15.6%)
Nivolumab	18 (23.4%)
Pembrolizumab	28 (36.4%)

Safety

Related Treatment Emergent Adverse Events Occurring in ≥ 5% of Participants attributed to mRNA-2752

	Arm B (N=77)			
	Gr 1 (%)	Gr 2 (%)	Gr 3* (%)	Total
Pyrexia	19 (24.7)	6 (7.8)	3 (3.9)	28 (36.4)
Injection site pain	8 (10.4)	9 (11.7)	5 (6.5)	22 (28.6)
Chills	12 (15.6)	4 (5.2)	0	16 (20.8)
Fatigue	10 (13.0)	4 (5.2)	1 (1.3)	15 (19.5)
Flushing	4 (5.2)	7 (9.1)	0	11 (14.3)
Injection site erythema	7 (9.1)	4 (5.2)	0	11 (14.3)
Nausea	8 (10.4)	2 (2.6)	0	10 (13.0)
Injection site swelling	5 (6.5)	3 (3.9)	0	8 (10.4)
Decreased appetite	5 (6.5)	0	0	5 (6.5)
Vomiting	2 (2.6)	3 (3.9)	0	5 (6.5)
Cytokine release syndrome	2 (2.6)	2 (2.6)	0	4 (5.2)
Diarrhoea	3 (3.9)	1 (1.3)	0	4 (5.2)
Hypotension	1 (1.3)	3 (3.9)	0	4 (5.2)
Injection site oedema	2 (2.6)	2 (2.6)	0	4 (5.2)
Myalgia	4 (5.2)	0	0	4 (5.2)
Tachycardia	1 (1.3)	3 (3.9)	0	4 (5.2)
Tumour pain	2 (2.6)	1 (1.3)	1 (1.3)	4 (5.2)

*Two events of Gr 3 tumor necrosis attributed to mRNA-2752, necessitating treatment interruption; both continued with durvalumab treatment without mRNA-2752

No Gr 4 or 5 AEs were attributed to mRNA-2752

Clinical Efficacy

Triple Negative Breast Cancer Expansion Cohort

Skin examination for TNBC patient (PD-L1+ low) demonstrating response in injected lesion:

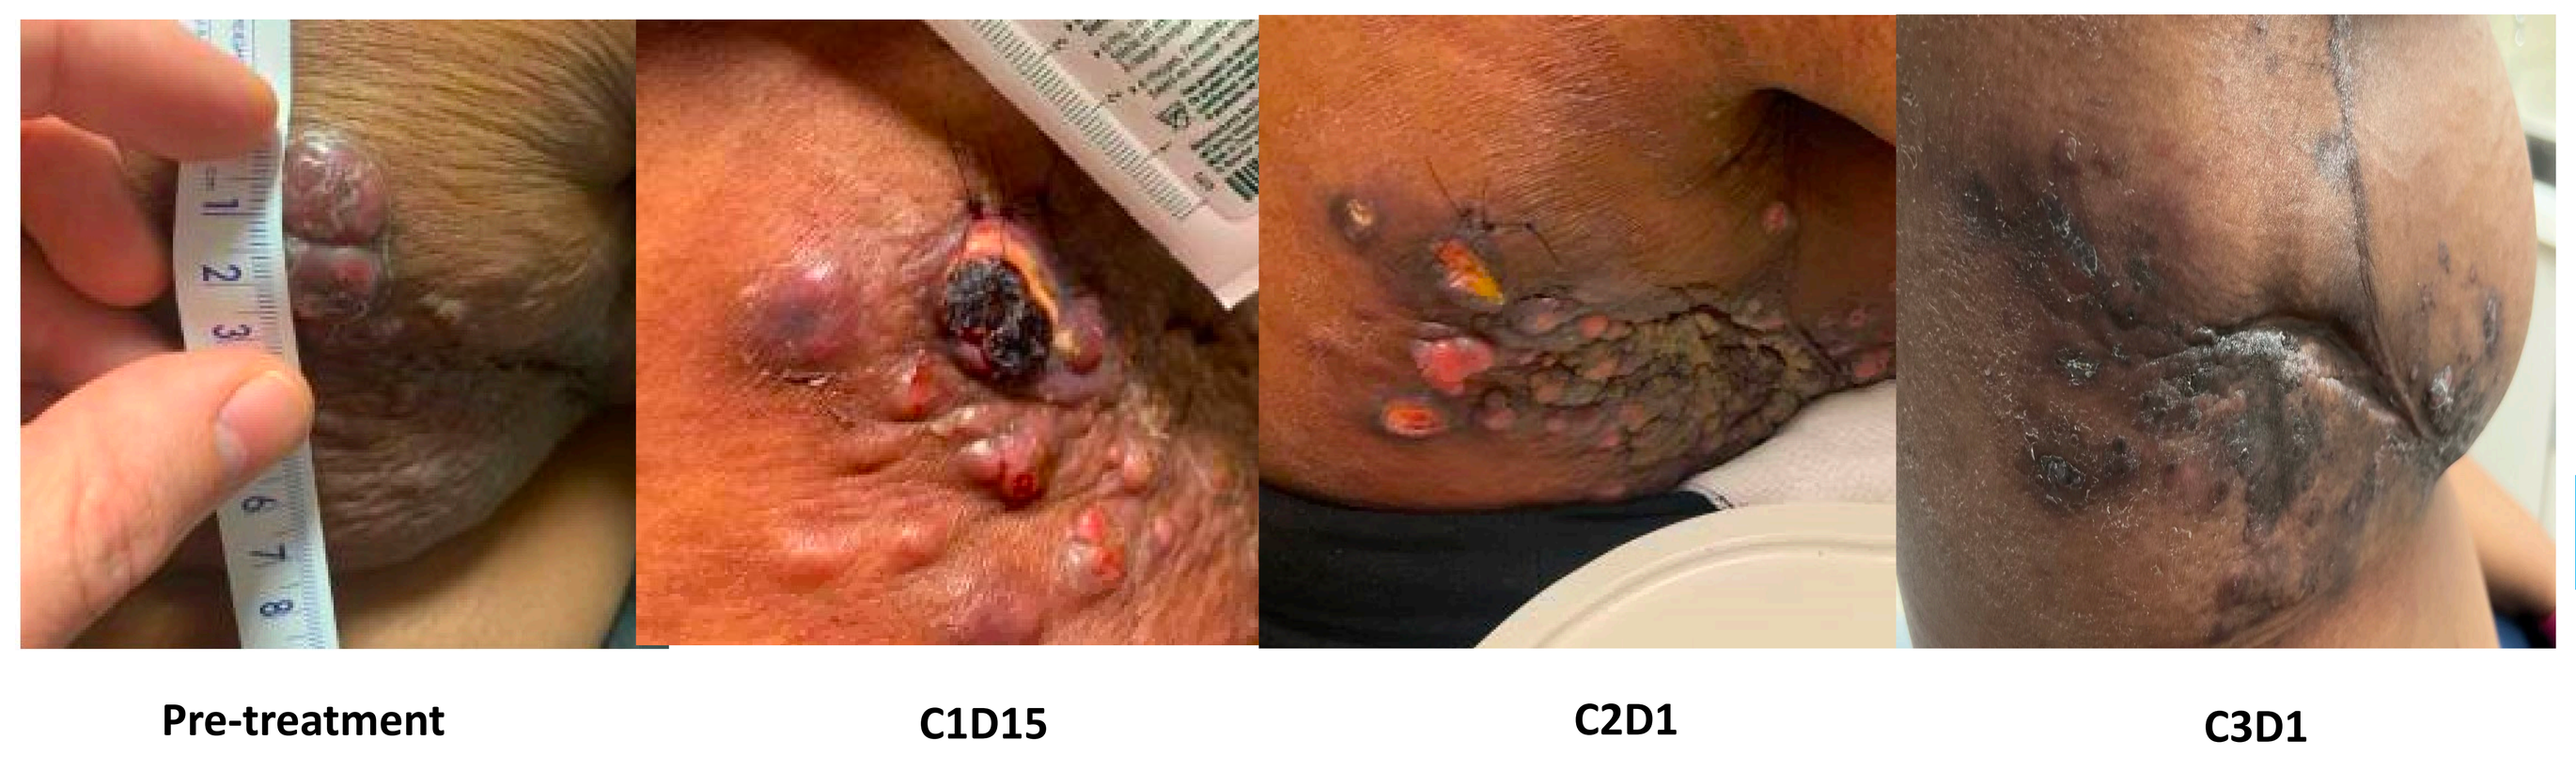

Pre-treatment | C1D15 | C2D1 | C3D1

TNBC patient, s/p R partial mastectomy, RT R breast and distant LN, progressed on 5 prior lines including Trop-2 ADC. Baseline PD-L1 scores were low: tumor cells = 0%; immune cells = 3%. Subcutaneous nodules became necrotic after the first injection with flattening and resolution of all surrounding satellite lesions with subsequent injections. On-treatment core biopsies of injected and satellite lesions showed no residual carcinoma. PR ongoing for 6+ months.

Percent Change from Baseline in Sum of Diameters of Target Lesions Over Time Based on Investigator Assessment per RECIST 1.1

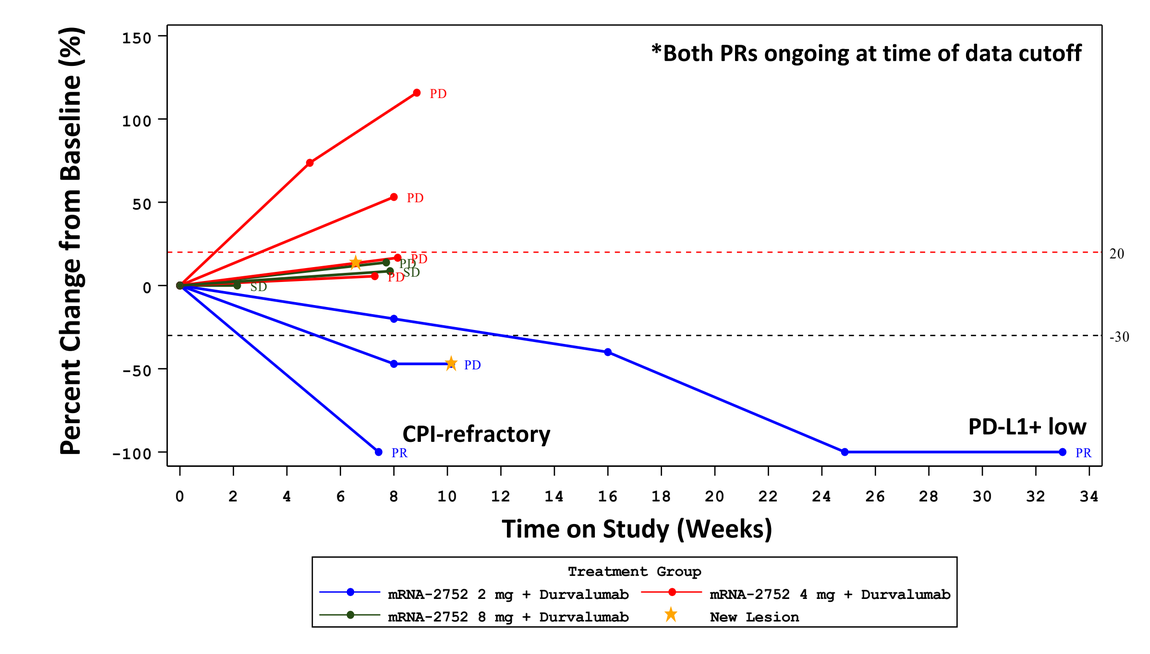

*Both PRs ongoing at time of data cutoff
Percent Change from Baseline (%)
CPI-refractory
PD-L1+ low
Time on Study (Weeks)
Treatment Group
mRNA-2752 2 mg + Durvalumab
mRNA-2752 4 mg + Durvalumab
mRNA-2752 8 mg + Durvalumab
New Lesion

Maximum %Change from Baseline in Sum of Diameters of Target Lesions by Injection Status Based on Investigator Assessment per RECIST 1.1

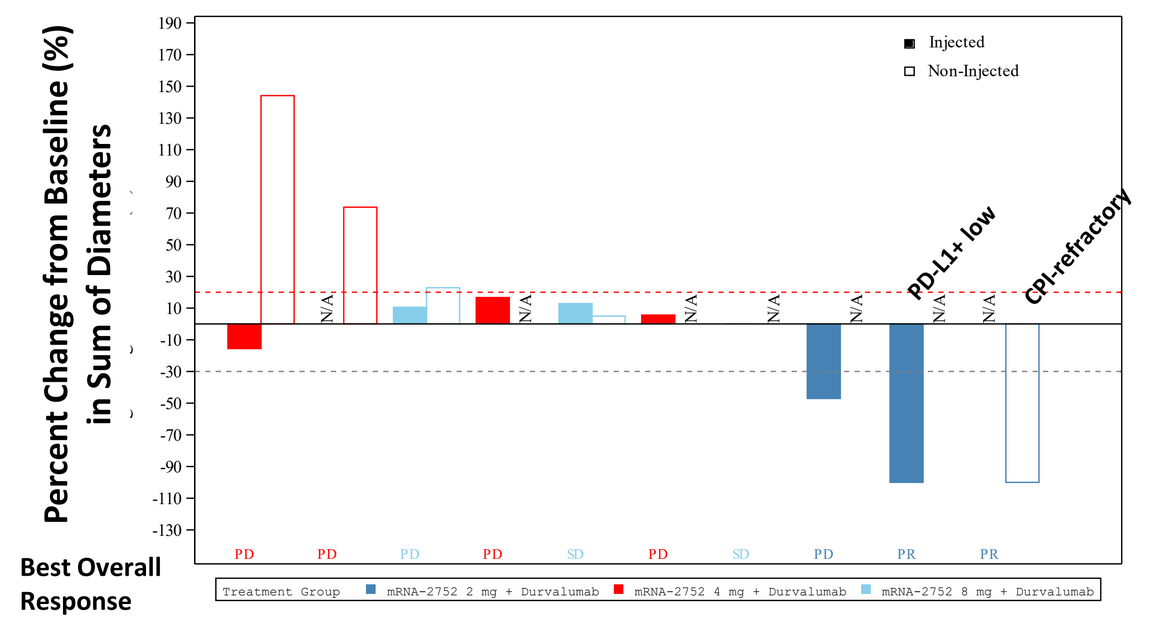

Injected
Non-Injected
PD-L1+ low
CPI-refractory
Percent Change from Baseline (%) in Sum of Diameters
Best Overall Response

Cutaneous Melanoma Expansion Cohort

Percent Change from Baseline in Sum of Diameters of Target Lesions Over Time Based on Investigator Assessment per RECIST 1.1

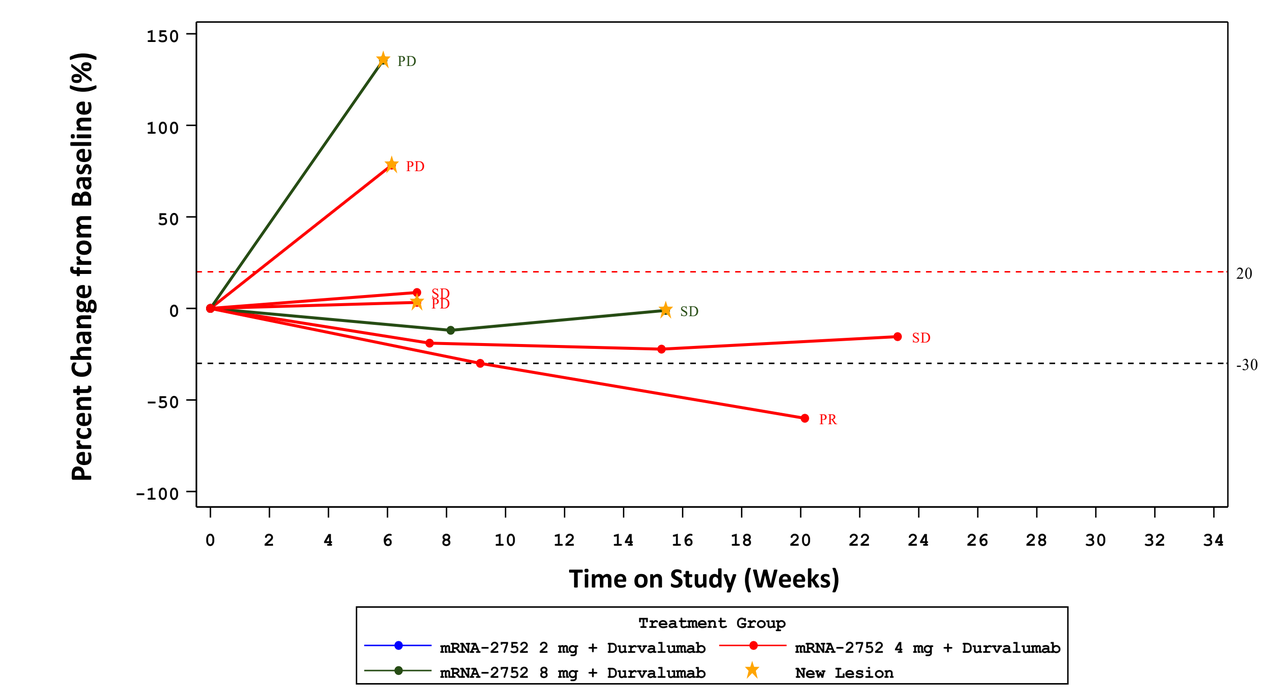

Percent Change from Baseline (%)
Time on Study (Weeks)
Treatment Group
mRNA-2752 2 mg + Durvalumab
mRNA-2752 4 mg + Durvalumab
mRNA-2752 8 mg + Durvalumab
New Lesion

Maximum %Change from Baseline in Sum of Diameters of Target Lesions by Injection Status Based on Investigator Assessment per RECIST 1.1

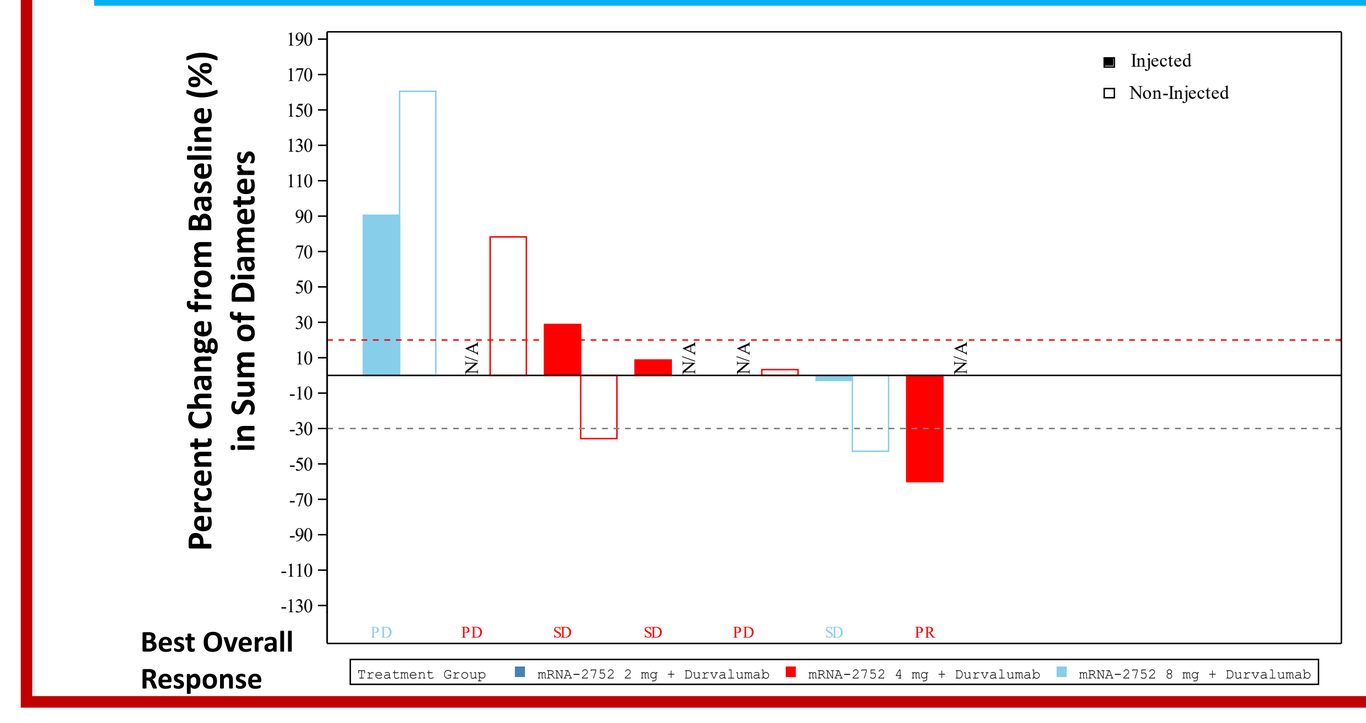

Injected
Non-Injected
Percent Change from Baseline (%) in Sum of Diameters
Best Overall Response

Radiologic Images for CPI-refractory cutaneous melanoma patient with complete resolution of non-injected level 5A LN (below) and lung metastases (not shown):

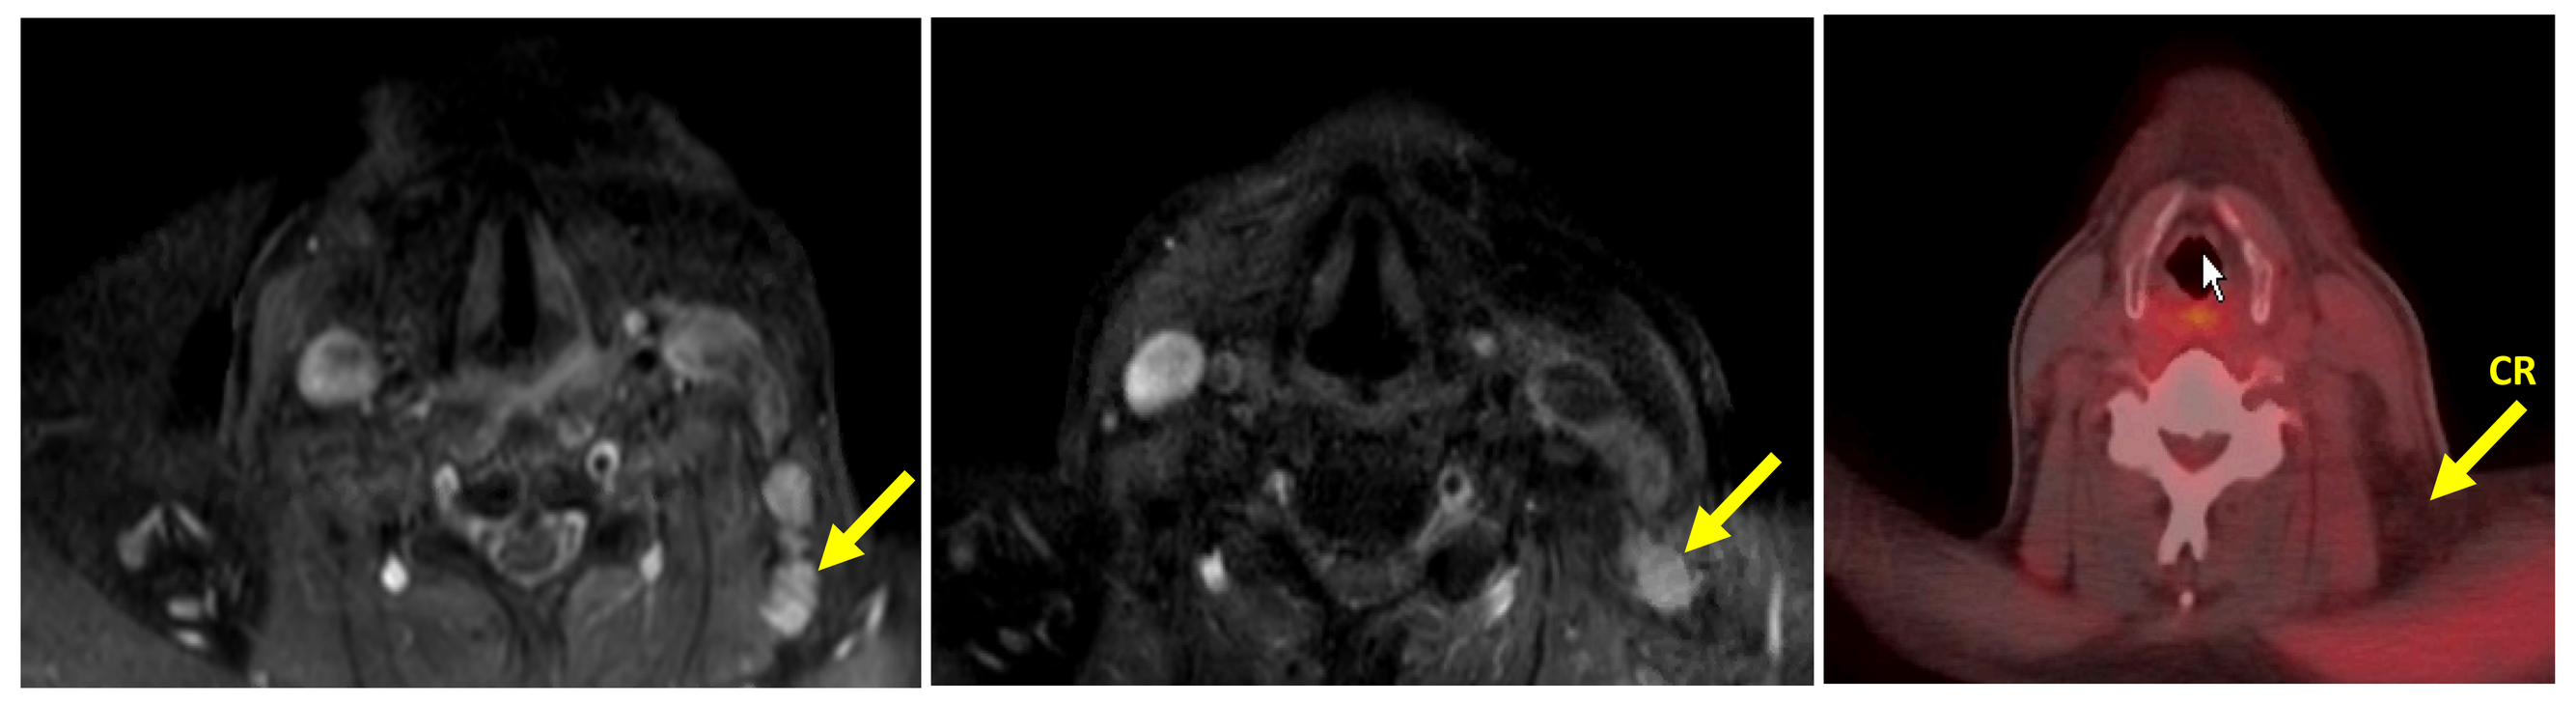

CR

Pre-treatment | C2D1-EOT | 7 month follow-up

Patient previously progressed on nivolumab/ ipilimumab and pembrolizumab; pseudoprogression of injected ear lesion noted at C2D1; confirmed iCR ongoing at 7 month follow-up.

Pharmacodynamics in the plasma

Intra-tumoral injection of mRNA-2752 induces expression of IL-23, IL-36γ and effector cytokines IFN-γ and TNF-α in the plasma

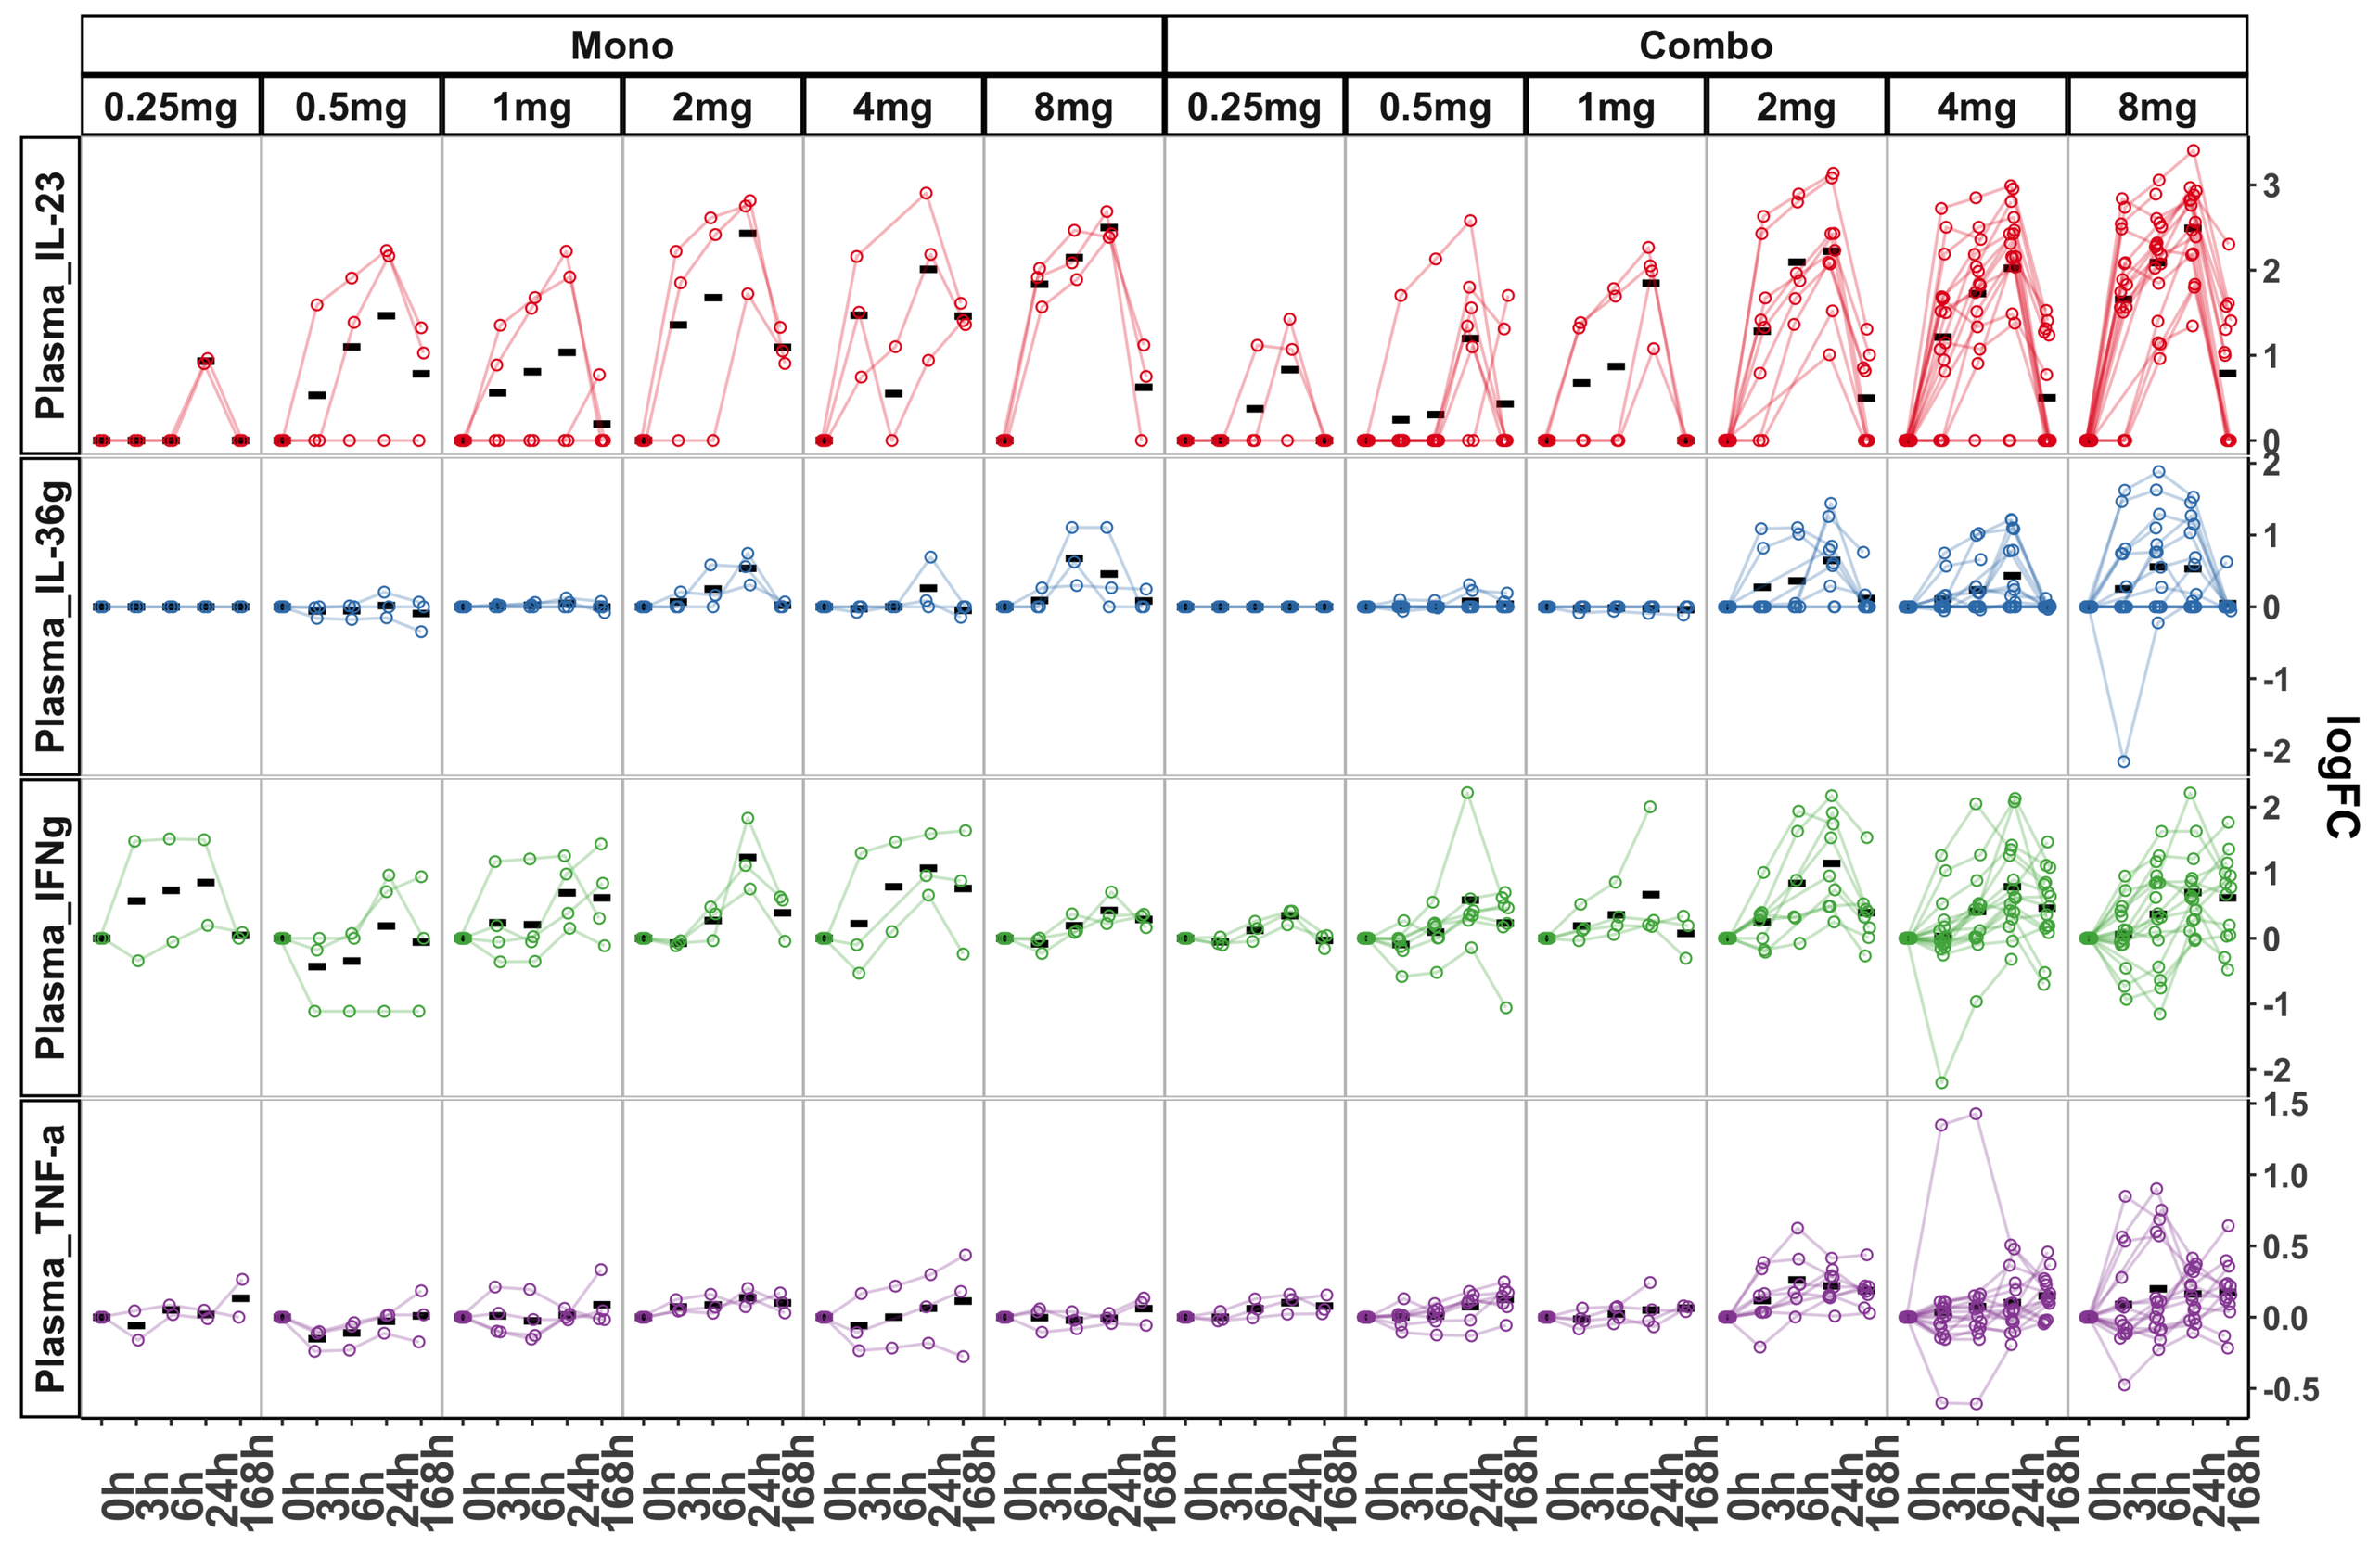

Mono
Combo
0.25mg
0.5mg
1mg
2mg
4mg
8mg
Plasma_IL-23
Plasma_IL-36g
Plasma_IFNg
Plasma_TNF-a
logFC
0h
3h
6h
24h
168h

Plasma cytokine were assessed by multi-plex MSD assay and IL-36γ ELISA. BLQ results are shown as LLOQ value / 2; for reported ALQ, samples were diluted and re-assessed whenever possible, or highest level detected for each analyte was plotted. Analysis of the fold changes relative to baseline after mRNA-2752 injection as monotherapy (mono) or in combination with durvalumab (combo) were assessed across the dose groups. mRNA-2752 alone induces effector cytokine production, and addition of durvalumab potentiates the effect with increased IFN-γ produced at 6- and 24-hours following injection (with descriptive p-value of 0.029 and 0.049 respectively without multiplicity adjustment).

Pharmacodynamics in the tumor microenvironment (TME)

mRNA-2752 alone induces expression of IL-23, IL-36γ, and effector cytokines IFNγ and TNFα at C1D2 and is sustained at C2D1 in combination with durvalumab

MONO

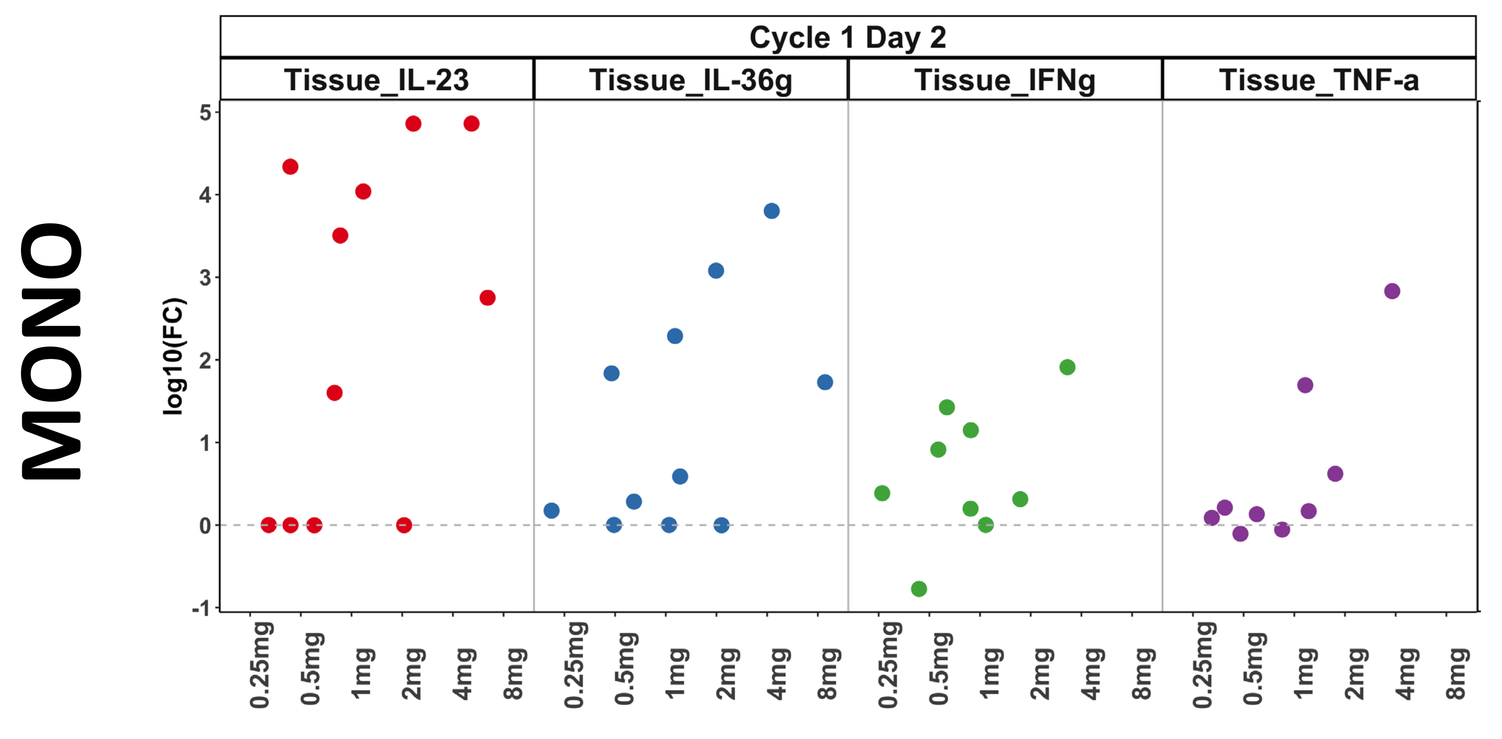

Cycle 1 Day 2
Tissue_IL-23
Tissue_IL-36g
Tissue_IFNg
Tissue_TNF-a
log10(FC)

Increased protein expression after mRNA-2752 injection monotherapy. Frozen tumor lysates were assessed similarly to plasma samples. Fold changes of intra-tumoral IL-23, IL-36γ (introduced by mRNA-2752) and effector cytokines are shown at C1D2 relative to baseline across dose levels.

COMBO

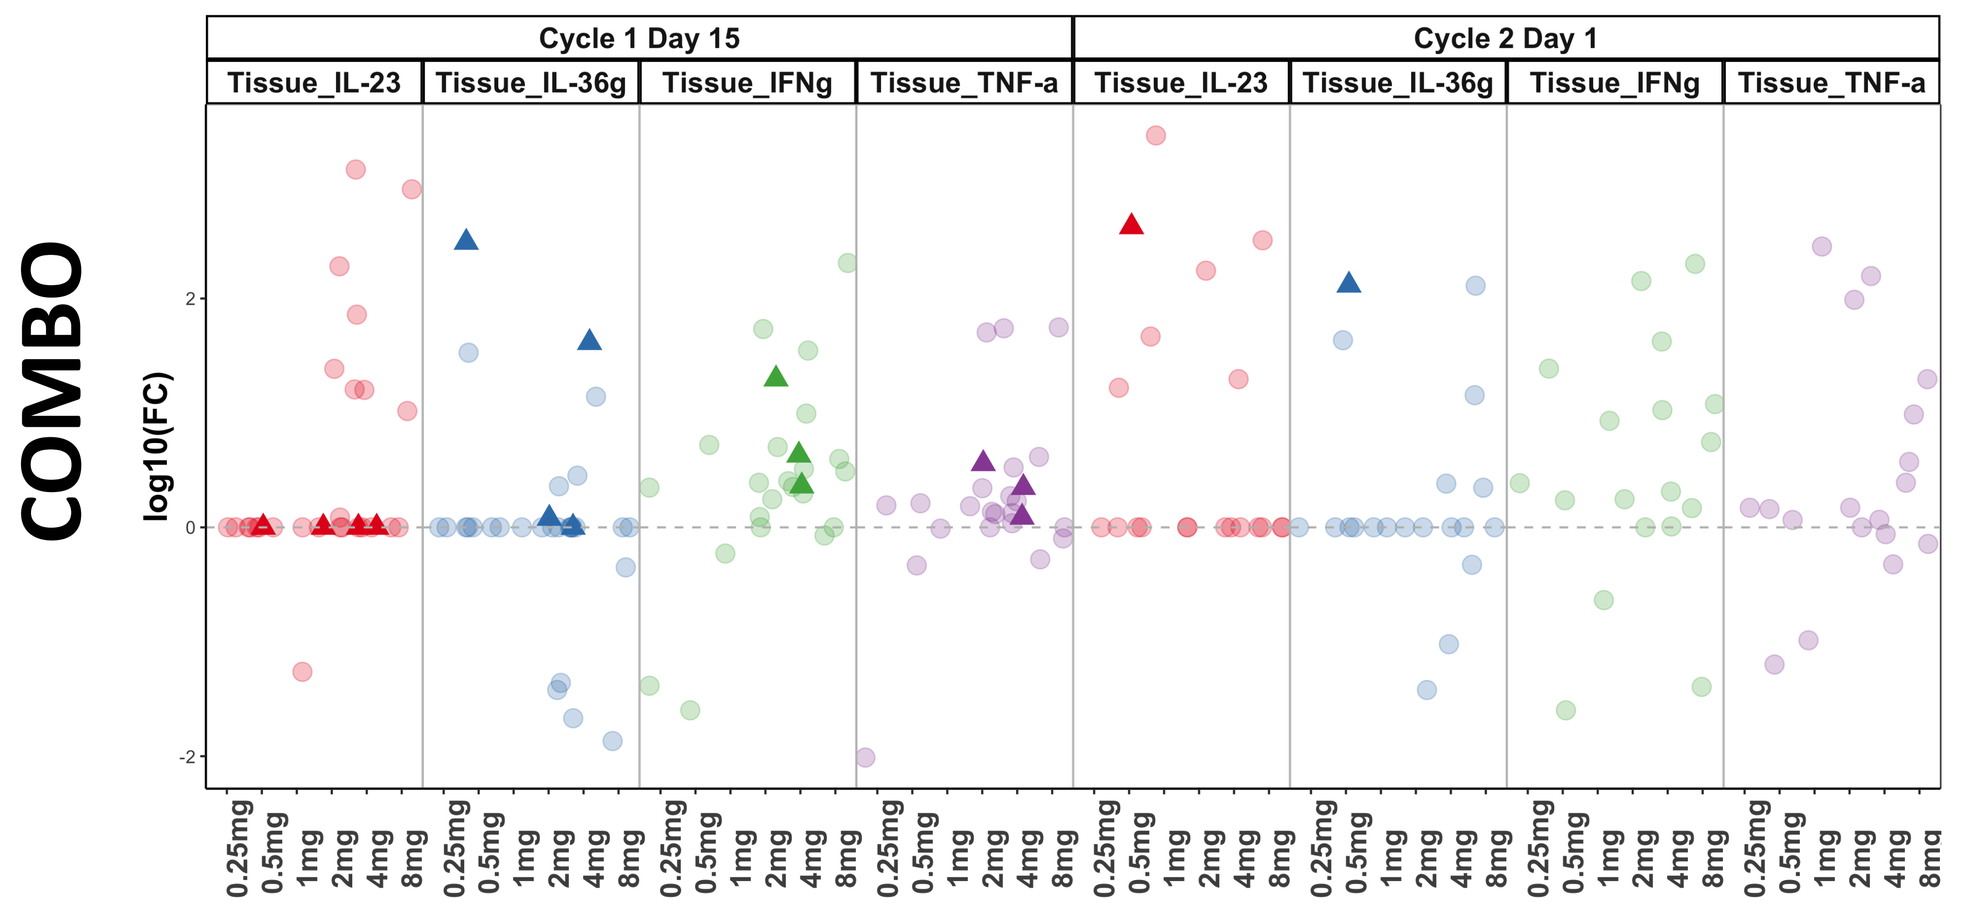

Cycle 1 Day 15
Cycle 2 Day 1
Tissue_IL-23
Tissue_IL-36g
Tissue_IFNg
Tissue_TNF-a
log10(FC)

Protein expression and effector response in combination therapy. Fold changes of intra-tumoral IL-23, IL-36γ, IFN-γ and TNF-α are shown at C1D15 and C2D1 relative to baseline, across all combination doses. Cytokine levels in responder patients (PR/CR) are noted with triangles.

mRNA-2752 in combination with durvalumab induces an inflammatory response in the TME, including signatures of increased innate and adaptive immune cell abundance and effector response

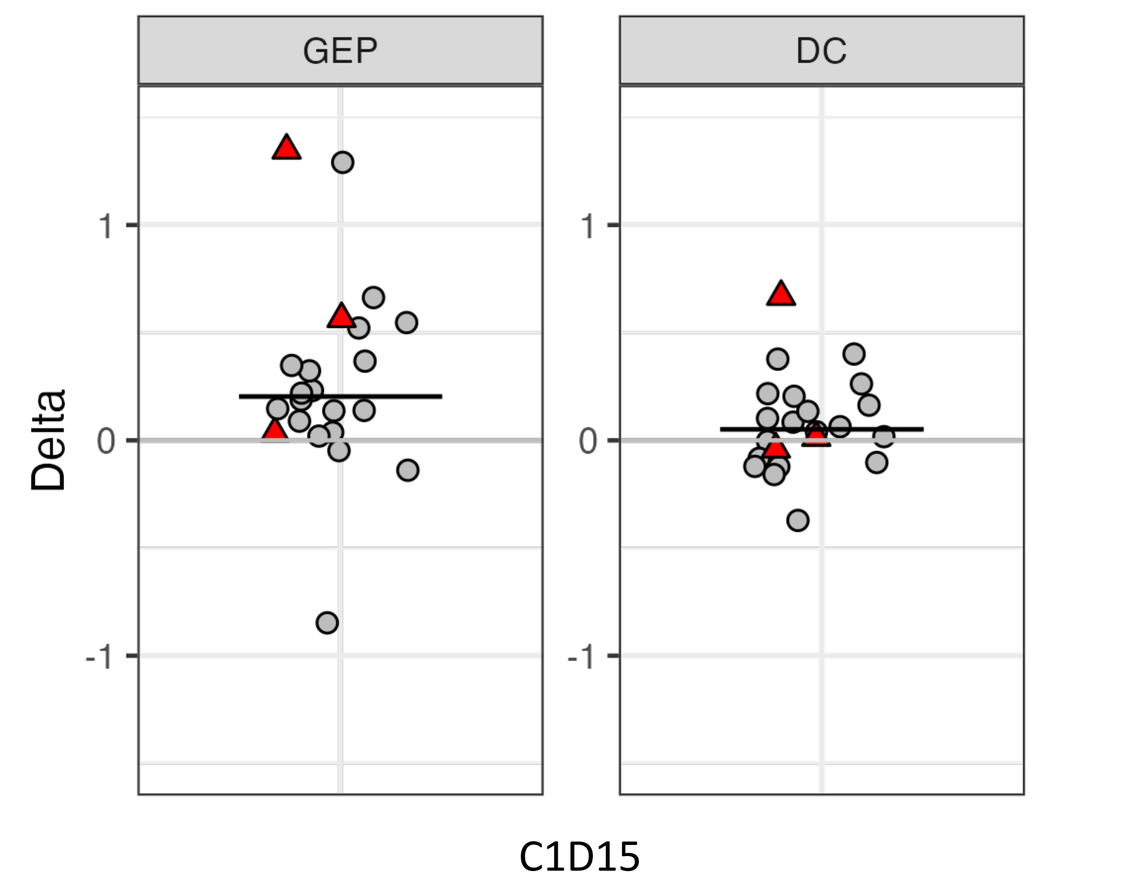

GEP
DC
Delta
C1D15

Increased DC activation and IFN signaling in the TME after treatment as measured by gene expression signatures. Change in T cell-inflamed (GEP score; Ayers et al.) and DC (Mayoux et al.) transcriptional signatures on-treatment relative to baseline. Combo patients were assessed at C1D15 and are shown; red triangles: PR/CR responders.

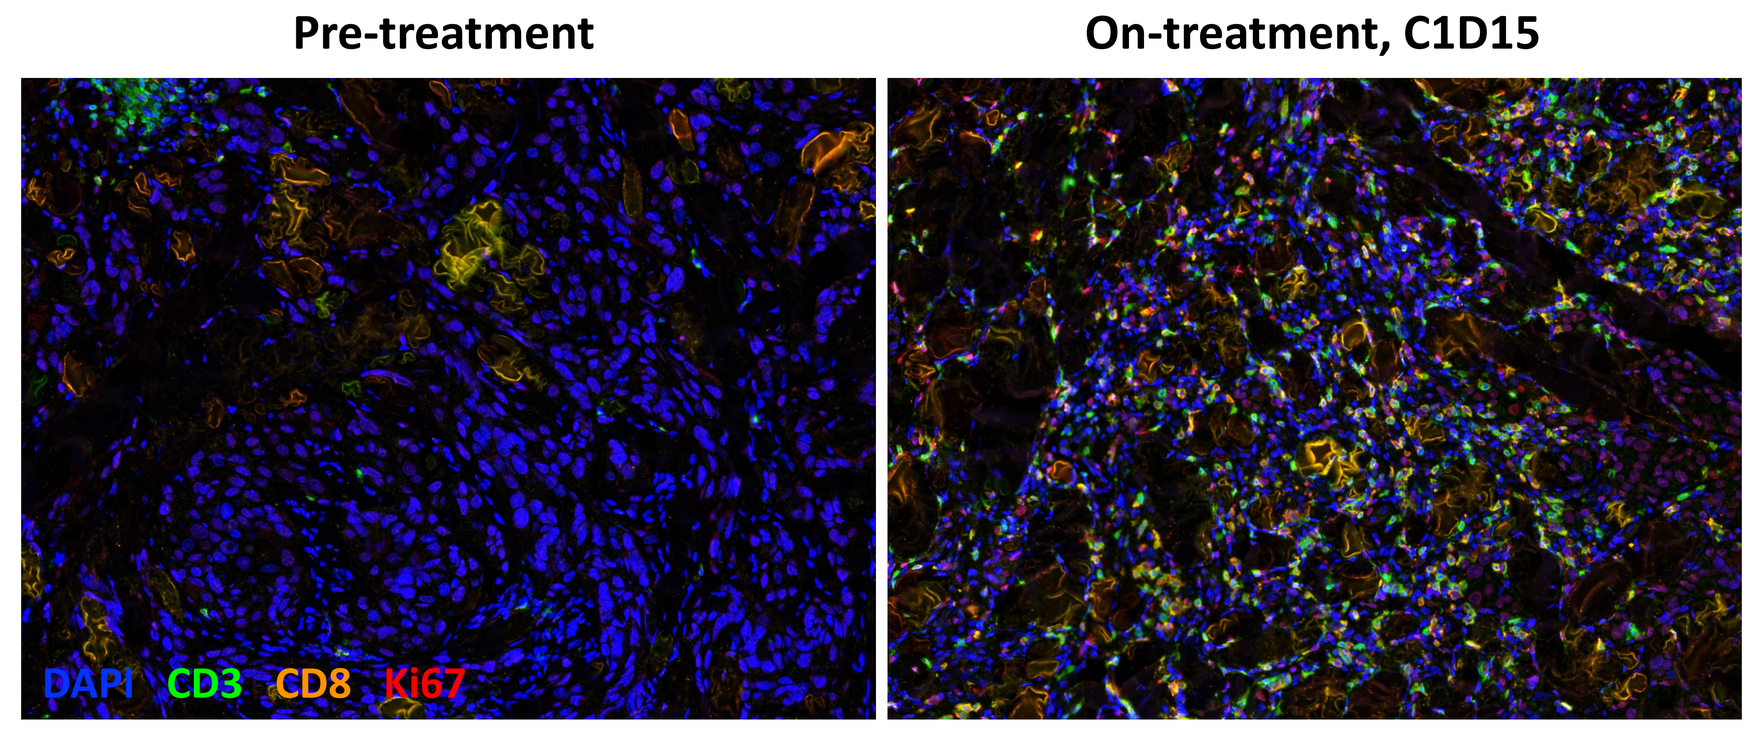

Pre-treatment
On-treatment, C1D15
DAPI CD3 CD8 Ki67

Ki67+ of CD8+: 36%; CD3+: 1% | Ki67+ of CD8+: 53%; CD3+: 7%

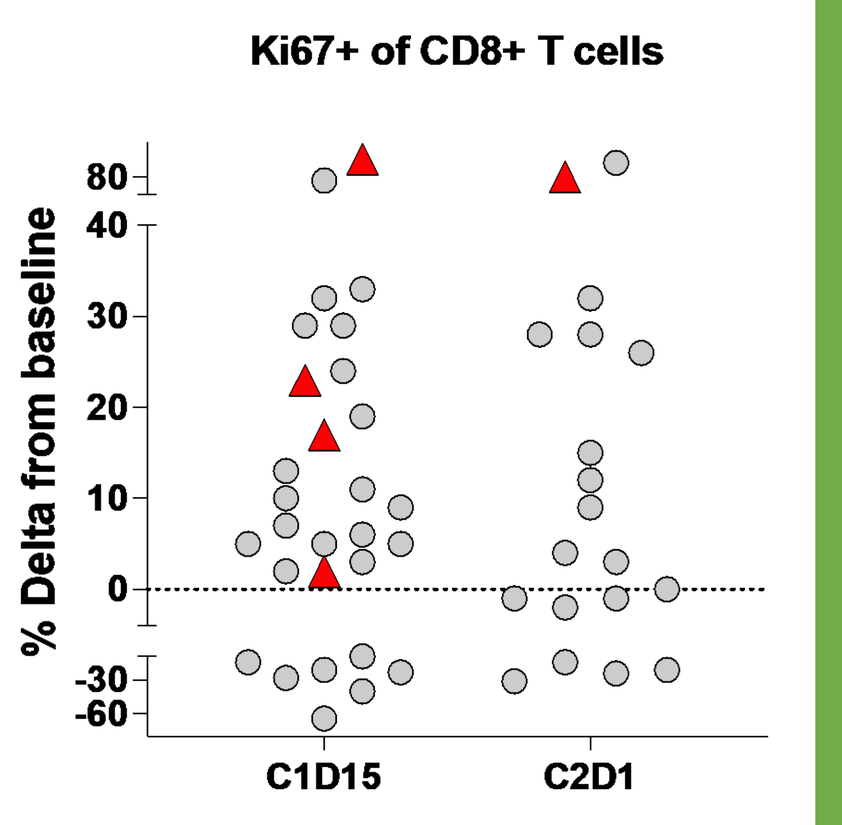

Ki67+ of CD8+ T cells
% Delta from baseline
C1D15
C2D1

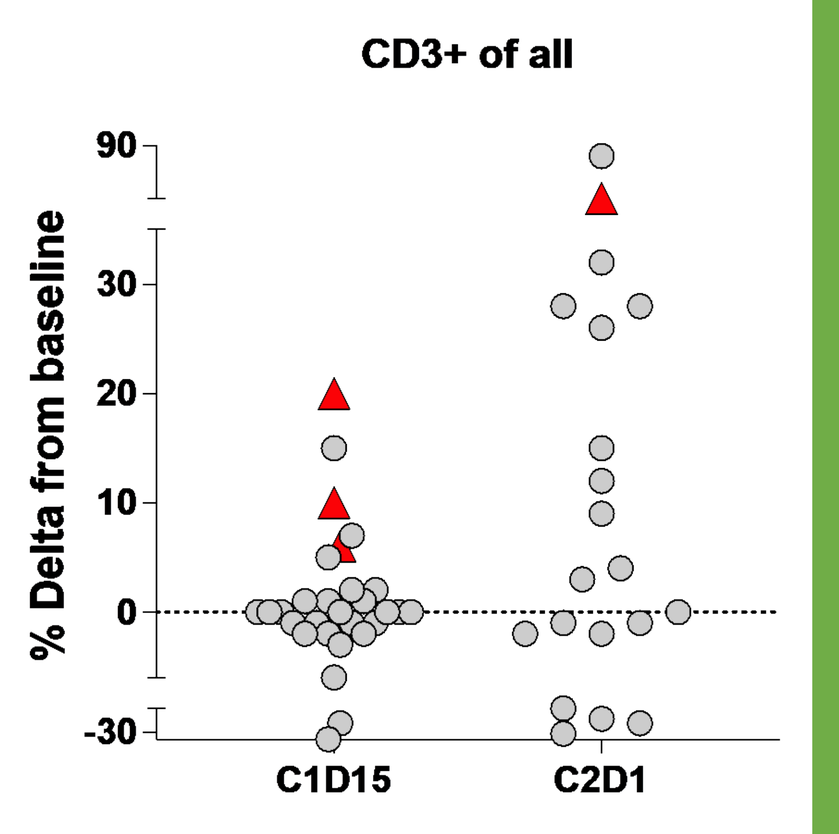

CD3+ of all
% Delta from baseline
C1D15
C2D1

Elevated TIL proliferation and abundance in the TME following treatment. Changes in percent of intra-tumoral Ki67+ of CD8+ T cells and overall CD3+ T cells are shown at C1D15 & C2D1 relative to baseline for combo patients. Representative F-IHC images are shown for TNBC patient (PR ongoing for 6+ months); red triangles: PR/CR responders.

CONCLUSIONS

- iTu mRNA-2752 up to 8mg + IV durvalumab is safe and tolerable
- ITu administration of mRNA-2752 in combination with durvalumab is associated with tumor shrinkage in both injected and un-injected lesions
- Clinical responses were seen in PD-L1 low and CPI-refractory TNBC, and CPI-refractory melanoma
- mRNA-2752 intra-tumoral injection as monotherapy or in combination with durvalumab induces encoded protein expression of IL-23, IL-36γ, as well as effector cytokines IFN-γ, TNF-α in the tumor and plasma
- Consistent with the expected mechanism of action, a sustained inflammatory response is observed in the TME, including increased immune cell abundance and signatures of increased innate and adaptive effector response

References

Hewitt et al. **Durable anticancer immunity from intratumoral administration of IL-23, IL-36γ, and OX40L mRNAs.** Sci Transl Med 2019.

Patel et al., **A phase I study of mRNA-2752, a lipid nanoparticle encapsulating mRNAs encoding human OX40L, IL-23, and IL-36γ, for intratumoral (iTu) injection alone and in combination with durvalumab.** J Clin Oncol 2020.

Ayres et al., **IFN-γ-related mRNA profile predicts clinical response to PD-1 blockade.** J Clin Invest 2017.

Cristescu et al., **Pan-tumor genomic biomarkers for PD-1 checkpoint blockade-based immunotherapy.** Science 2018.

Mayoux et al., **Dendritic cells dictate responses to PD-L1 blockade cancer immunotherapy.** Sci Transl Med 2020.

Acknowledgements

All patients and their families
AstraZeneca for provision of durvalumab

NCT03739931